Cumulative Oral Corticosteroid Use Increases Risk of Glucocorticoid-related Adverse Events in Patients with Pemphigus

David Wormser, PhD[1*]; Diana M. Chen, MD[2]; Paul G. Brunetta, MD[2]; Michael S. Broder, MD[3]; Eunice Chang, PhD[3]; Sheila R. Reddy, PhD[3]

[1] F. Hoffman-La Roche Ltd, Basel, Switzerland; [2] Genentech, Inc., South San Francisco, CA, USA; [3] Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA
** david.wormser@roche.com*

INTRODUCTION

- Pemphigus is a group of chronic autoimmune disorders that is characterized by intraepithelial blistering of the skin and mucous membranes that can be life-threatening.
- The only FDA-approved treatment for patients with pemphigus is corticosteroid (CS) therapy, which acts to inhibit blister formation and allow healing of erosions by way of immunosuppression.
- While mortality rates due to pemphigus have declined substantially with the introduction of corticosteroids, there is significant morbidity, including serious and fatal complications, associated with high dose and prolonged CS exposure in the treatment of pemphigus.
- To date there has been no reported evidence on the estimated risk of glucocorticoid-related adverse events (GAE) in pemphigus that could better inform the clinical management of pemphigus, which may include the use of unapproved adjuvant therapies to minimize CS exposure.

OBJECTIVES

- The main purpose of this study was to investigate the risk of GAE among pemphigus patients taking oral corticosteroids (OCS).

METHODS

Study Design

- We analyzed commercial and Medicare supplemental claims data retrospectively (2010-2014) to examine risk and number of GAE and associated costs among patients ≥ 18 years of age with:
 - A newly diagnosed episode of pemphigus defined as: ≥ 2 claims in the identification period with an ICD-9-CM code of 694.4 (includes pemphigus vulgaris, foliaceus, and paraneoplastic variants) and with no pemphigus diagnosis within 6 months before the first date of diagnosis (index date).
 - Patients were required to have ≥ 1 claim for an OCS (per National Drug Code, NDC) within the 6 months before (baseline period) or the 6 months after the index date. We considered baseline OCS use to account for possible empirical steroid treatment used prior to a confirmed diagnosis of pemphigus.
- Patients were continuously enrolled during the baseline period through the first year of follow-up. After the first-year, follow-up time varied among patients (the median observation time was 785 days) **(Figure 1)**.

Figure 1. Study Time Frame

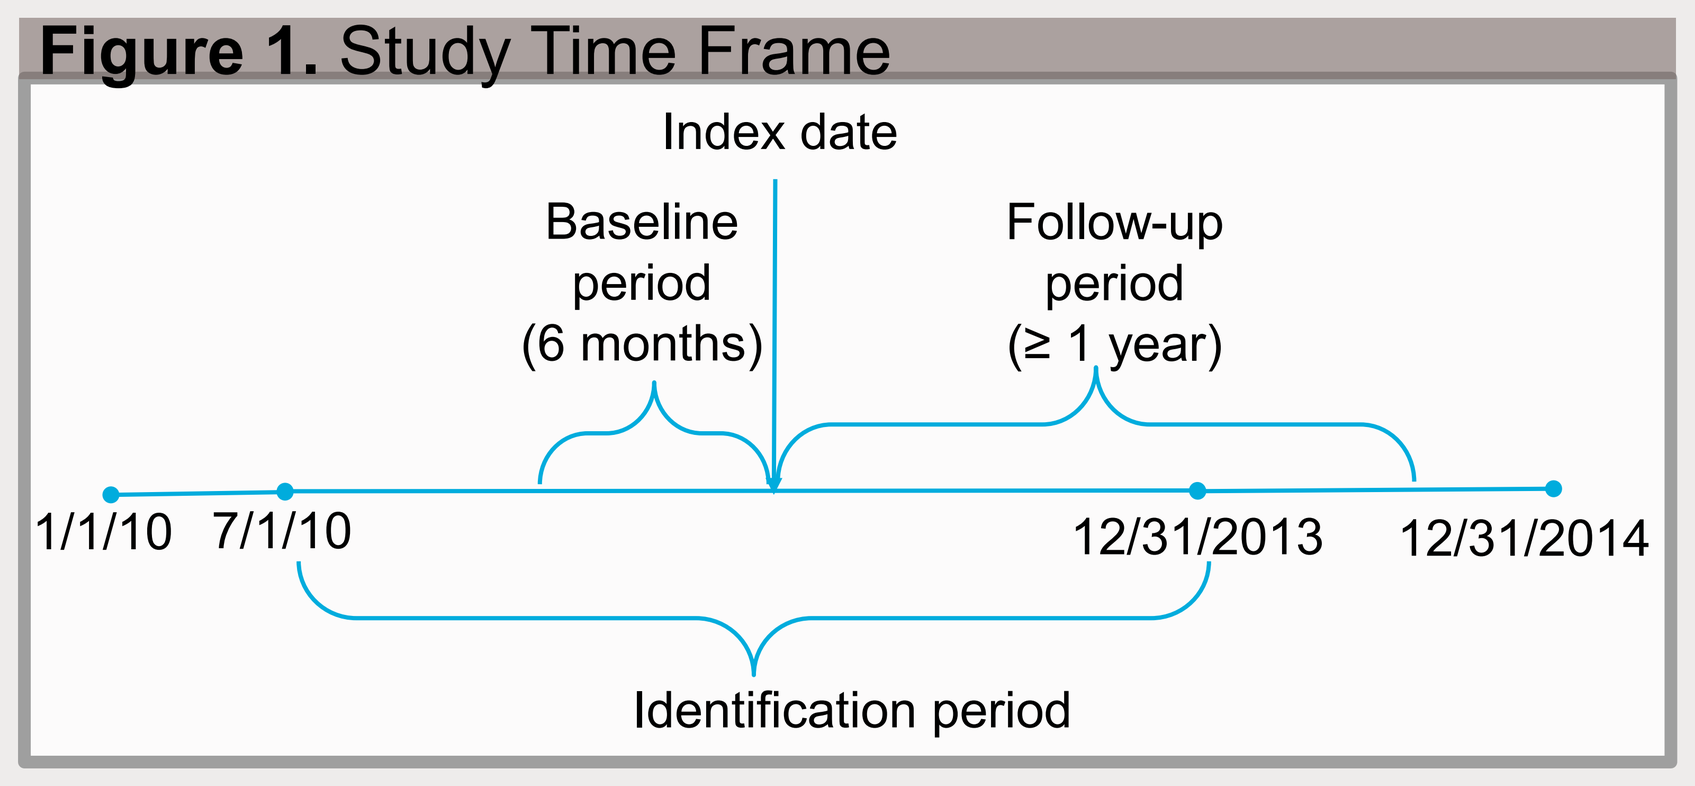
Index date
Baseline period (6 months)
Follow-up period (≥ 1 year)
1/1/10
7/1/10
12/31/2013
12/31/2014
Identification period

Measures

- Baseline measures included age, sex, geographic region, comorbidities, insurance plan type, OCS (prednisone-equivalent) dose and days supply, and number of chronic conditions (counted using the Agency for Healthcare Research and Quality (AHRQ) Healthcare Cost and Utilization Project (HCUP) Chronic Condition Indicator[1,2]).
- Main predictor of *cumulative OCS exposure* was constructed as a time-variant measure of cumulative OCS dose received from the beginning of the baseline period and updated daily (daily aggregated prednisone-equivalent dose).[3]
 - OCS dose was calculated using the NDC (to determine strength) and the quantity and days supplied information in the pharmacy claims.
- Primary outcome of *risk of GAE* was measured as the presence of a GAE *(yes/no)* during the follow-up period using International Classification of Diseases, Ninth Revision (ICD-9-CM) and Current Procedural Terminology (CPT) codes, overall and by category.
 - Chronic GAE were considered events if they did not occur during the baseline period. All acute GAE were considered events.
 - Chronic GAE: cardiovascular disease, cataract, glaucoma, osteoporosis, type 2 diabetes, and hypertension.
 - Acute GAE: opportunistic infections (serious), aseptic necrosis of bone, fractures, gastrointestinal ulcer, and psychosis.
- Secondary outcomes included *number of GAE* and *healthcare costs.*
 - Number of GAE was measured as the overall count of GAE during follow-up.
 - For cardiovascular disease, pulmonary embolism, myocardial infarction, stroke, and heart failure were counted as separate events.
 - For opportunistic infections, each infection was counted separately.
 - Overall, medical, and pharmacy costs were measured by totaling the paid amount (fee-for-service equivalent) in each claim over the first year of follow-up.

Statistical Analysis

- We used multivariate Cox regression models to estimate risk of GAE associated with cumulative OCS exposure during ≥1 year follow-up. We investigated the interaction term of OCS exposure and event time and it was not significant, indicating no evidence for rejecting proportionality.
- Multivariate negative binomial regression was used to examine the relationship between cumulative OCS exposure and the number (or rate) of GAE.
- First-year costs were compared between patients with and without GAE (in the first year) using analysis of covariance (ANCOVA).
- All statistical models were adjusted for age, gender, and comorbidity.
- All data processing and analyses were performed using SAS® version 9.4.

RESULTS

Baseline Characteristics

- We identified 644 patients with a new pemphigus episode (**Figure 2**). Mean age was 59.5 years (SD=16.9), 56.1% were female, 43.9% were male, and patients came from all geographic regions within the United States.
- Nearly two-thirds (64.6%) of patients had commercial insurance; 35.4% had an employer-sponsored Medicare Supplemental plan **(Table 1)**.
- Patients had a mean Charlson Comorbidity Index of 0.9 (SD=1.6), and on average 2.7 chronic conditions (SD=2.2) at baseline **(Table 1)**.
- 10.1% of patients had another autoimmune condition: type 1 diabetes (6.1%), rheumatoid arthritis (2.3%), inflammatory bowel disease (1.9%), autoimmune thyroid disease (0.3%), and myasthenia gravis (0.2%) **(Table 1)**.
- Total dose and days supply of OCS in the first follow-up year was 827.1mg (median=210mg, interquartile range (IQR, 25th-75th percentile) was 0.0-871mg) and 35.3 days (median=10 days; IQR=0.0-56.5mg), respectively. The corresponding average daily dose of OCS was 19.0mg (median=11.9mg; IQR=0.0-25.0mg) (**Table 1**).

Figure 2. Cohort Identification: Patients with Pemphigus

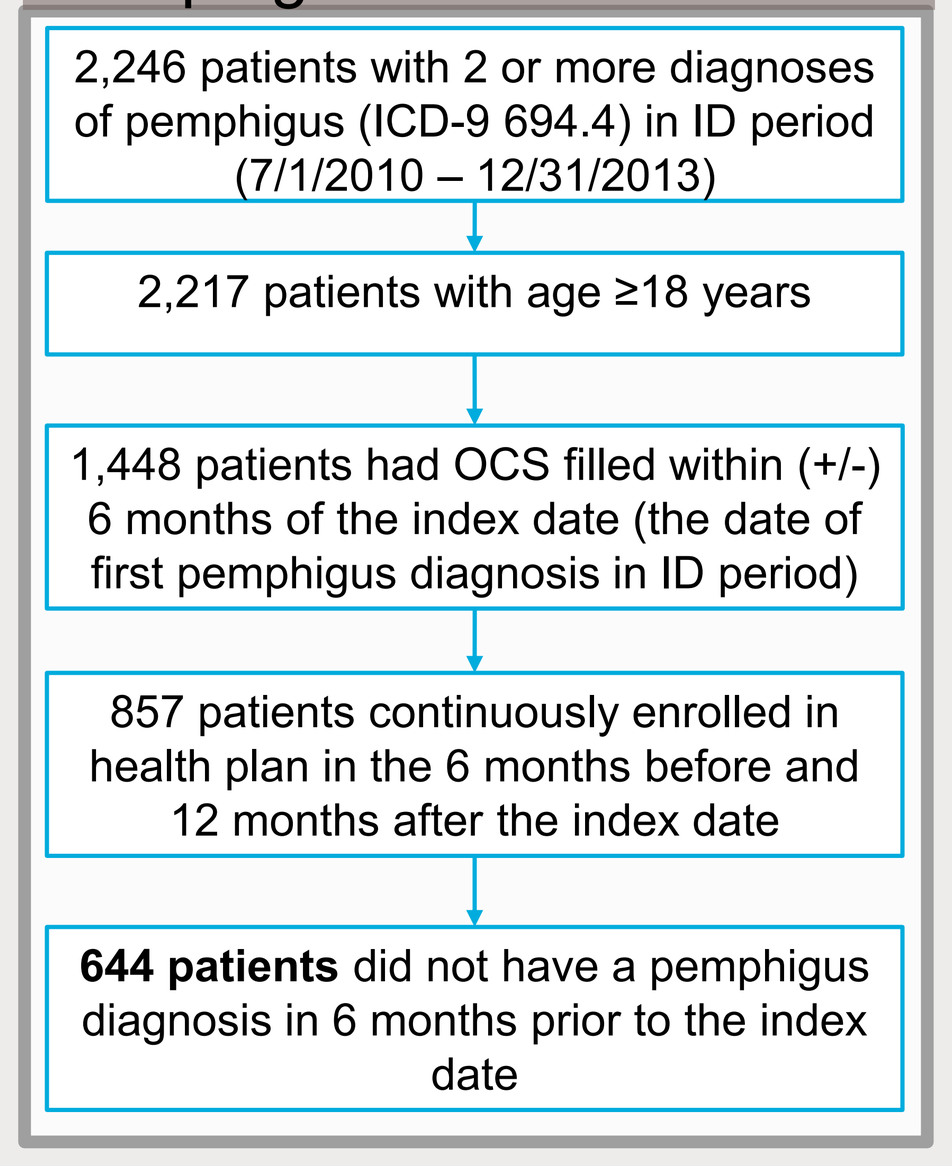
2,246 patients with 2 or more diagnoses of pemphigus (ICD-9 694.4) in ID period (7/1/2010 – 12/31/2013)
2,217 patients with age ≥18 years
1,448 patients had OCS filled within (+/-) 6 months of the index date (the date of first pemphigus diagnosis in ID period)
857 patients continuously enrolled in health plan in the 6 months before and 12 months after the index date
644 patients did not have a pemphigus diagnosis in 6 months prior to the index date

Table 1. Baseline Characteristics [a]

	All N=644
Age, year, mean (SD)	59.5 (16.9)
18-49, n (%)	190 (29.5)
50-59	145 (22.5)
60-69	119 (18.5)
70+	190 (29.5)
Gender, n (%)	
Female	361 (56.1)
Male	283 (43.9)
US Region, n (%)	
North Central	129 (20.0)
Northeast	157 (24.4)
South	235 (36.5)
West	123 (19.1)
Plan Type, n (%)	
Commercial	416 (64.6)
Medicare supplemental [b]	228 (35.4)
Charlson Comorbidity Index, mean (SD)	0.9 (1.6)
No. chronic conditions, mean (SD) [c]	2.7 (2.2)
Other Autoimmune conditions, n (%)	65 (10.1)
Myasthenia gravis	1 (0.2)
Autoimmune thyroid disease	2 (0.3)
Inflammatory bowel disease	12 (1.9)
Rheumatoid arthritis	15 (2.3)
Type 1 diabetes	39 (6.1)
Total dose of OCS, mg, mean (SD) [median] [d]	827.1 (2,388.3) [210.0]
Total days of supply of OCS, mean (SD) [median] [e]	35.3 (48.1) [10.0]
Average daily dose of OCS, mg, mean (SD) [median] [f]	19.0 (35.2) [11.9]

[a] Baseline characteristics measured during 6 months prior to the index date. [b] Medicare-eligible retirees with employer-sponsored Medicare Supplemental plans. [c] Chronic Condition Indicator developed by the AHRQ HCUP. [d] Interquartile range (IQR, 25th-75th percentile) was 0.0-871.0mg. [e] IQR was 0.0-56.5.0mg. [f] IQR was 0.0-25.0mg.

Risk and Number of GAE during Follow-up

- Cox regression estimated the hazard ratio (HR) per 1g OCS exposure to be 1.01 (P=0.03), that is, each additional 1g of OCS was associated with a 1% higher risk of having a GAE **(Table 2)**.
 - Risk of cataract (HR: 1.02; P<0.001) and fracture (HR: 1.01; P=0.03) also increased with OCS exposure **(Table 2)**.
 - An observed increase in risk of infection, osteoporosis, necrosis of bone, psychosis, glaucoma, and type 2 diabetes was not statistically significant (results not shown).

Table 2. Association between OCS Exposure and Risk of GAE [a,b,c]

	Risk of GAE		Risk of Cataract		Risk of Fractures	
	HR (95% CI)	*P* Value	HR (95% CI)	*P* Value	HR (95% CI)	*P* Value
Age, year						
50-59 vs. 18-49	2.33 (1.68 - 3.23)	<.001	2.41 (1.18 - 4.92)	0.016	1.77 (0.70 - 4.50)	0.231
60-69 vs. 18-49	2.95 (2.11 - 4.13)	<.001	4.71 (2.40 - 9.22)	<.001	3.31 (1.37 - 8.01)	0.008
70+ vs. 18-49	4.40 (3.18 - 6.09)	<.001	4.29 (2.21 - 8.33)	<.001	5.53 (2.45 - 12.46)	<.001
Female vs. Male	1.07 (0.87 - 1.31)	0.515	1.27 (0.88 - 1.83)	0.203	1.83 (1.11 - 3.03)	0.018
No. chronic conditions [d]	1.00 (0.95 - 1.05)	0.998	1.11 (1.02 - 1.21)	0.012	1.02 (0.91 - 1.14)	0.735
Cumulative exposure, per 1g prednisone-equivalent dose	1.01 (1.00 - 1.02)	0.029	1.02 (1.01 - 1.03)	<.001	1.01 (1.00 - 1.03)	0.034

[a] Cumulative OCS exposure measured since the beginning of 6-month baseline period and updated daily during follow-up. [b] Cox proportional hazard regression used to study the relationship between OCS exposure and GAE risk. Sample size for each model was 644. [c] An increased risk of infection, osteoporosis, necrosis of bone, psychosis, glaucoma, and type 2 diabetes was observed but not statistically significant. [d] Chronic Condition Indicator developed by the AHRQ HCUP.

- The unadjusted incidence rate of GAE was 0.53 events per patient year. In our adjusted model, the incidence rate ratio for GAE associated with cumulative OCS exposure was 1.02 (P<0.0001) **(Table 3)**, meaning each additional gram of OCS exposure was associated with a 2% increase in the number of GAE per year.

Table 3. Association between OCS Exposure and Number of GAE Per Year [a,b]

	IRR (95% CI)	*P* Value
Age, year		
50-59 vs. 18-49	2.14 (1.63 - 2.82)	<.0001
60-69 vs. 18-49	2.71 (2.05 - 3.59)	<.0001
70+ vs. 18-49	3.82 (2.94 - 4.96)	<.0001
Female vs Male	1.09 (0.92 - 1.28)	0.3231
No. chronic conditions [c]	1.01 (0.97 - 1.05)	0.8030
Cumulative exposure per year, per 1g prednisone-equivalent dose	1.02 (1.01 - 1.04)	<.0001

[a] Annualized cumulative OCS exposure measured since the beginning of 6-month baseline period until the end of follow-up. [b] Negative binomial regression used to test association between OCS exposure and number of GAE. Sample size for the model was 644. [c] Chronic Condition Indicator developed by the AHRQ HCUP.

First-Year Follow-up Costs

- Presence of GAE was associated with higher first-year overall and medical costs, but not outpatient pharmacy costs **(Figure 3)**.
 - In the first-year of follow-up, overall healthcare costs for patients with GAE exceeded those of patients without GAE ($46,722 vs. $24,589; P<0.001) **(Figure 3)**.
 - Patients with GAE also had higher mean medical costs ($42,198 vs. $21,004; P<0.001) **(Figure 3)**.
 - Outpatient pharmacy costs for patients with GAE were not statistically different than costs for patients without GAE ($4,524 vs. $3,585; P=0.090) **(Figure 3)**.

Figure 3. Adjusted First-Year Costs by GAE Status [a,b]

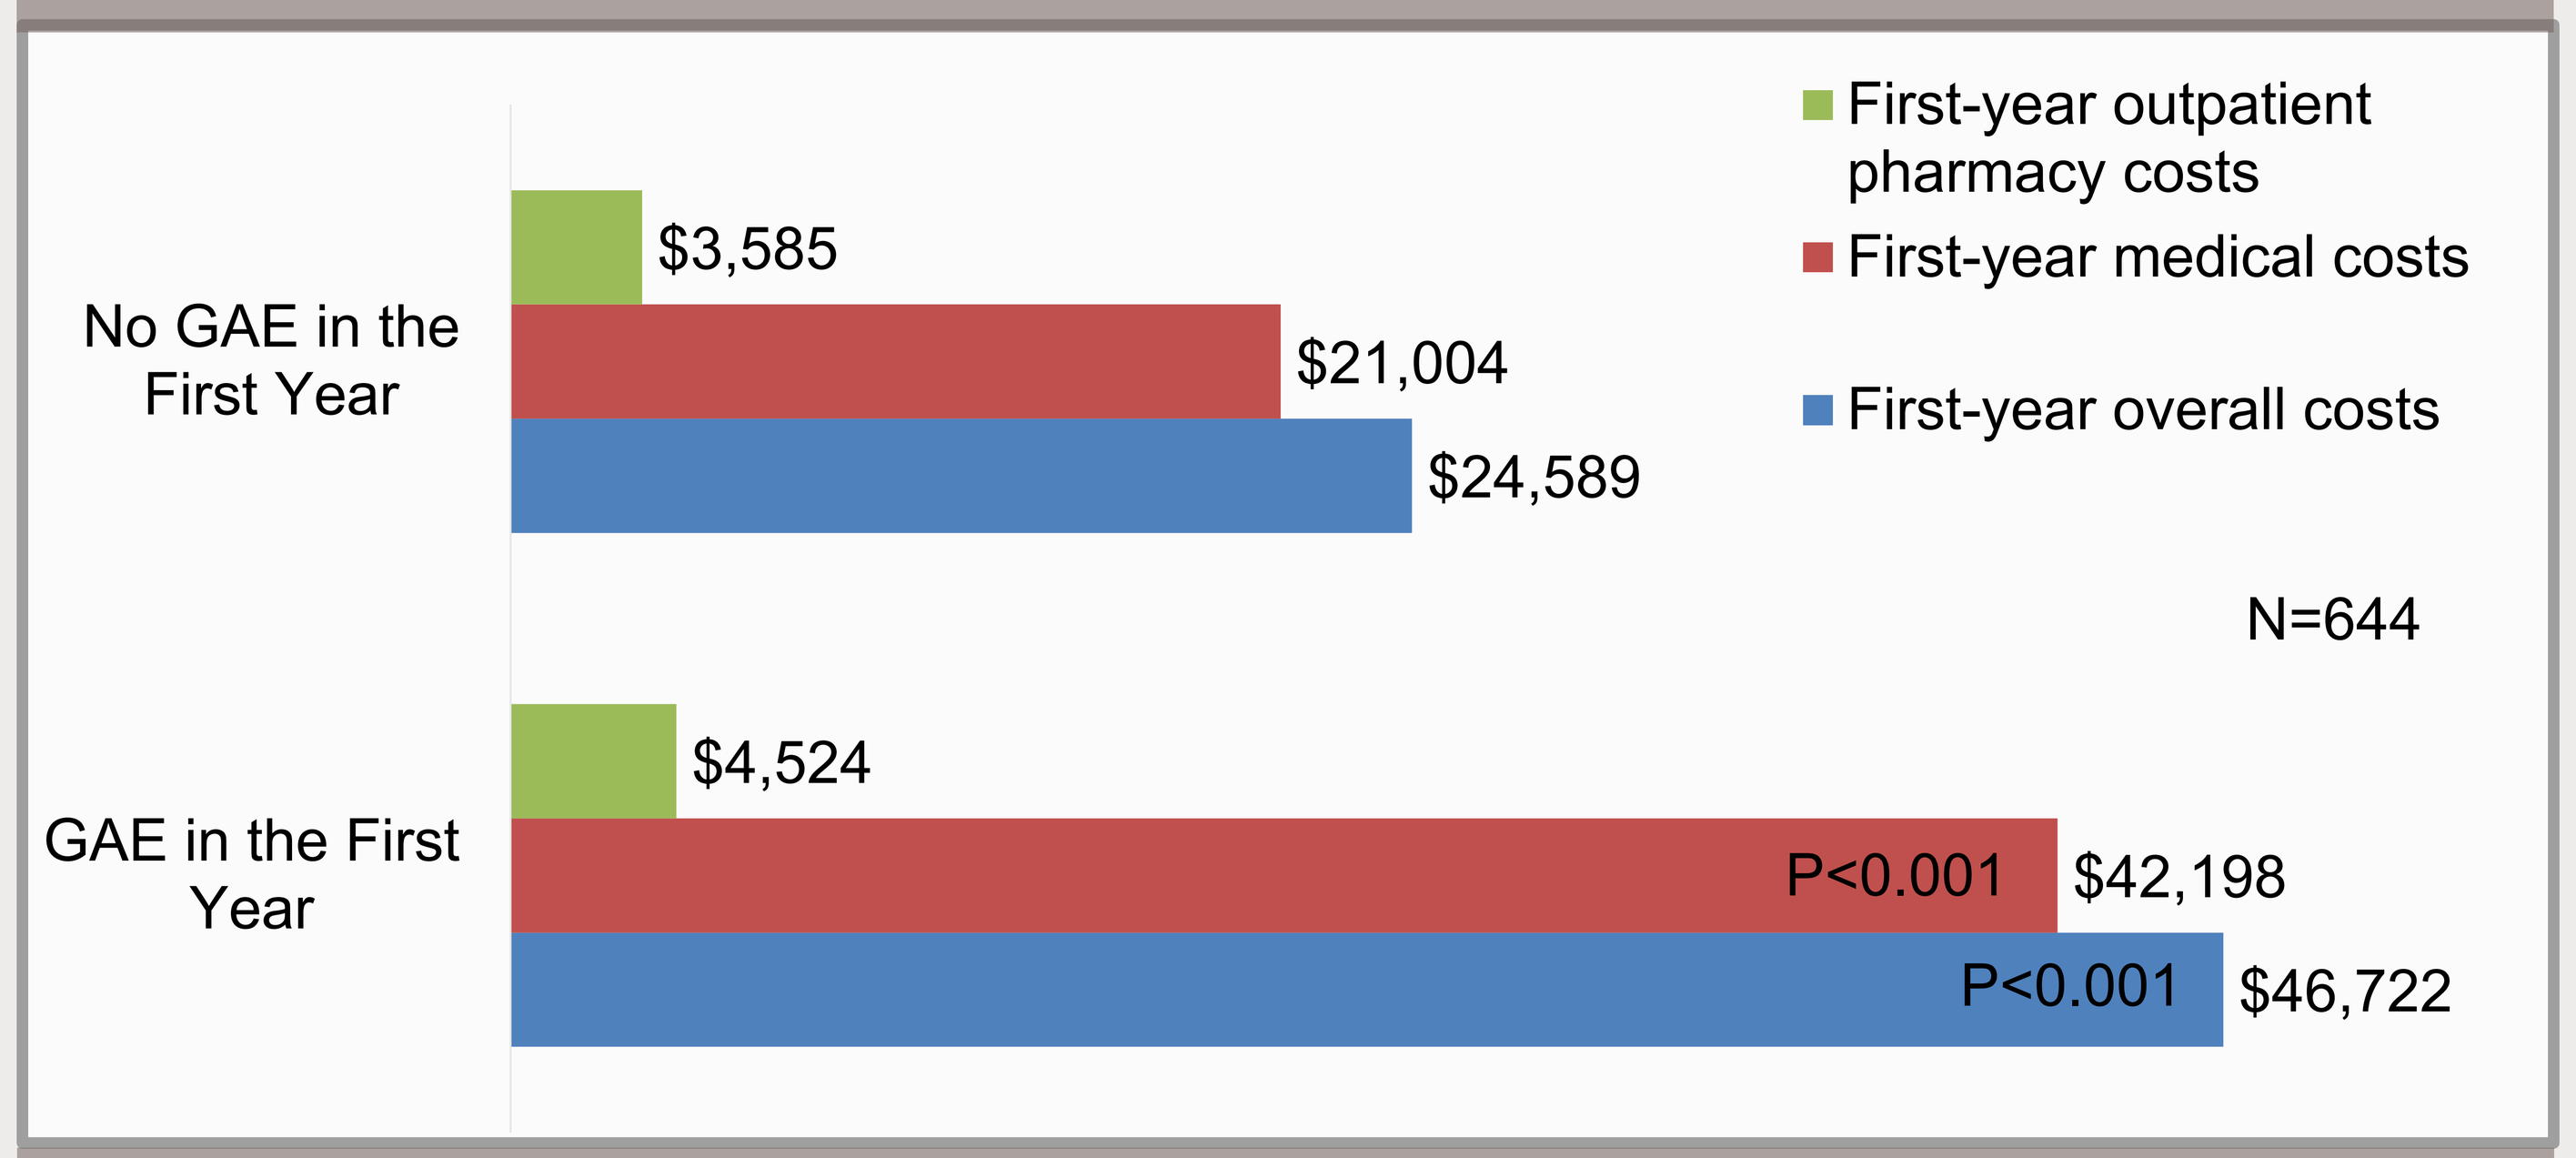
First-year outpatient pharmacy costs
First-year medical costs
First-year overall costs
No GAE in the First Year
$3,585
$21,004
$24,589
N=644
GAE in the First Year
$4,524
P<0.001
$42,198
P<0.001
$46,722

[a] First year following the index date. [b] Adjusted by age, gender, no. of chronic conditions.

LIMITATIONS

- Our analysis was based on OCS dispensing data as reported on pharmacy claims, which may not reflect how the medication was actually used.
- These results may not be generalizable to uninsured individuals or to those with other types of insurance (e.g., Medicaid) not included in this database.
- Administrative claims data are not collected or maintained for research purposes; thus, claims-based clinical information (e.g., ICD-9-CM codes associated with pemphigus or adverse events) does not contain the same degree of precision as in clinical studies, leading to possible over or underreporting of cases.

CONCLUSIONS

- This study demonstrates that pemphigus patients exposed to high cumulative OCS dose have a higher risk of cataract and fracture and that patients with GAE incur greater healthcare costs.
- Increasing OCS exposure is also associated with a greater number of GAE experienced per patient.
- The observed increase in risk for certain GAE may not have been statistically significant for several reasons, including prophylactic treatment for serious infections, such as pneumocystis, and osteoporosis.
- Clinical management focused on steroid-free remissions or lower OCS exposure may reduce the detrimental effects demonstrated in this study.
- Prospective evaluation of steroid-related toxicity in selected future clinical studies, using a validated glucocorticoid toxicity index, may help to further elucidate and weigh the burden of adverse events associated with corticosteroid therapy in pemphigus.[4]

REFERENCES

1. Agency for Healthcare Research and Quality. HCUP Chronic Condition Indicator. http://www.hcup-us.ahrq.gov/toolssoftware/chronic/chronic.jsp.
2. Hwang W et al. Health Affairs 2001;20(6):267-278
3. Garber, E. *Drugs for rheumatic disease* (eds. Paulus, H., Furst, D. & Dromgoole) 443 (Churchill Livingstone, 1987).
4. Development of a Glucocorticoid Toxicity Index (GTI) Using Multi-Criteria Decision Analysis. Manuscript under review.

Presented at the Society for Investigative Dermatology Annual Meeting, May 11-14, 2016, Scottsdale, AZ.

This study was funded by Roche.
Support for third-party writing assistance was provided by F. Hoffmann-La Roche Ltd.