Relationship Between HEDIS Measurements and Asthma-Related Outcomes

Karina Raimundo,[1] Paul G. Solari,[1] Michael Broder,[2] Eunice Chang,[2] James L. Zazzali[1]

[1]Genentech, Inc., South San Francisco, CA, USA; [2]Partnership of Health and Analytic Research, Beverly Hills, CA, USA

ABSTRACT

Background: The Healthcare Effectiveness Data and Information Set (HEDIS) asthma medication ratio (AMR) is a new quality of care indicator for patients with persistent asthma. Patients with a ratio <0.5 have been shown to be at higher risk for of asthma-related emergency department (ED) visits and hospitalizations. However, the risk of other asthma-related events is not well understood.

Objective: This study aims to evaluate how the AMR is associated with multiple indicators of poor asthma control.

Methods/Description: A retrospective cohort study was conducted using a commercial health care administrative claims database. Patients were identified as having persistent asthma, per the HEDIS definition, if they met any 1 of the following criteria in both 2010 and 2011: ≥1 ED visit (493.xx primary diagnosis); ≥1 hospitalization (493.xx primary diagnosis); ≥4 outpatient asthma visits and ≥2 asthma medication dispensing events; ≥4 asthma medication dispensing events. If a leukotriene modifier was the only asthma medication dispensed in that year, the patient was also required to have ≥1 diagnosis of asthma, in any setting, in the same year the leukotriene modifier was dispensed. Using data from 2011, AMR was calculated as the ratio of controller to total asthma medications. Patients with an AMR <0.5 were defined as low AMR. Poor asthma control events were defined as asthma-related hospitalizations, asthma-related ED visits, ≥2 oral corticosteroid (OCS) bursts, and ≥6 short-acting beta agonist (SABA) fills in a year. The number of asthma-related office visits in a year also was reported.

Results: A total of 37,054 patients met the study criteria and 6,479 (17.5%) had low AMR. Patients with low AMR had higher rates of any poor asthma control event compared with those with high AMR (74.3% vs 26.9%, respectively). Hospitalization rates (6.8% vs 1.9%, respectively), ED visits (24% vs 5.1%, respectively), ≥2 OCS bursts (32.0% vs 20.3%, respectively), and ≥6 SABA fills (49.2% vs 6.2%, respectively). The number of asthma-related office visits was higher for patients with low AMR (1.9 [standard deviation (SD), 2.0] vs 1.8 [SD, 1.9], respectively; *P*<0.001 for all comparisons).

Conclusions: Patients with low AMR (<0.5) had worse asthma control by multiple measures including hospitalization/ED visits, OCS use, and SABA use. The number of asthma-related office visits also was slightly higher for low AMR patients. The findings support the use of AMR as a quality of care measurement for patients with persistent asthma.

INTRODUCTION

- The Healthcare Effectiveness Data and Information Set (HEDIS) was developed by the National Committee for Quality Assurance and has been applied to many diseases, including asthma.[1]
- HEDIS is designed to:
 - Measure the effectiveness and satisfaction of delivered care and service;
 - Measure access to care; and
 - Measure how well the organization equips its members to make informed choices about their health.
- The HEDIS asthma measures identify patients with persistent asthma based on medical (inpatient and outpatient) and pharmacy claims data and assess the effectiveness of care through 3 separate measures: appropriate use of medications, medication management, and asthma medication ratio (AMR).
- The AMR identifies patients at high risk for poor asthma outcomes. Patients with a ratio <0.5 are at higher risk of asthma-related emergency department (ED) visits and hospitalization;[2] however, the risk of other asthma-related events is not well understood.

OBJECTIVE

- This analysis aimed to assess the value of the AMR as a measure of quality control by evaluating how the ratio is associated with multiple indicators of poor asthma control.

METHODS

Study Population and Time Frame

- A retrospective cohort study was conducted using medical and pharmacy data from a commercial health care administrative claims database. 2010 was the premeasurement year and 2011 was the measurement year (**Figure 1**).

Figure 1. Study Design

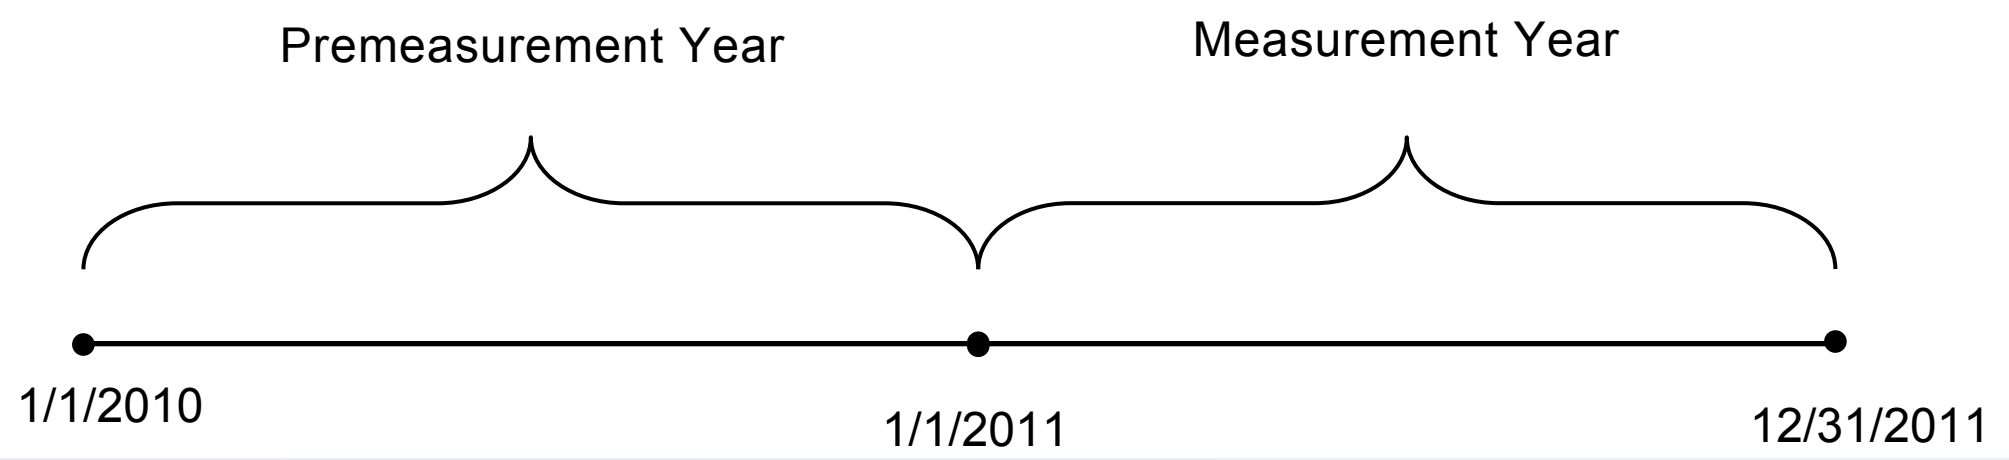
Premeasurement Year
Measurement Year
1/1/2010
1/1/2011
12/31/2011

Inclusion Criteria

- ≥1 primary diagnosis of asthma (International Classification of Diseases, Ninth Revision, Clinical Modification [ICD-9-CM] code 493.xx) in both the premeasurement year and measurement year.
- Commercial member with an age of 5–64 years by December 31, 2011.
- ≤1 gap in enrollment of ≤45 days during both the premeasurement year and measurement year.
- Per the HEDIS definition, patients had persistent asthma (ICD-9-CM code 493.xx) if they met any 1 of the following 4 criteria in both the premeasurement year and measurement year:
 - ≥1 ED visit(s) with asthma as the primary diagnosis;
 - ≥1 hospitalization with asthma as the primary diagnosis;
 - ≥4 outpatient asthma visits and ≥2 asthma medication dispensing events; and
 - ≥4 asthma medication dispensing events.
 - If leukotriene modifiers were the sole asthma medication dispensed in that year, the patient also was required to have ≥1 diagnosis of asthma, in any setting, in the same year the leukotriene modifier was dispensed.

AMR Calculation

- Using data from 2011 (the measurement year), the AMR was calculated using the following formula:

$$AMR = \frac{\textit{Units of controller medications}}{\textit{Units of controller + reliever medications}}$$

- Patients with an AMR <0.5 were defined as having a low AMR.

Asthma Outcomes

- Poor asthma control events were defined as the following:
 - ≥6 short-acting beta agonist (SABA) fills in a year;
 - ≥2 oral corticosteroid (OCS) bursts (≤15-day supply);
 - Asthma-related hospitalizations; and
 - Asthma-related ED visits.
 - Asthma-related ED visits were defined as claims in which the primary diagnosis code was for asthma (ICD-9-CM code 493.xx).
- The number of asthma-related office visits in a year also was reported.
 - Office visits were defined as evaluation and management visits and included office or other outpatient services, office consultations, and preventive medicine services.

Statistical Analysis

- All data transformations and statistical analyses were performed using SAS® version 9.4 (SAS Institute Inc., Cary, NC). Descriptive statistics, including means, standard deviations (SD), medians, and percentages, were reported for all measures whenever applicable.
- To compare the high and low AMR groups, chi-square tests and *t* tests were used for categorical and continuous variables, respectively. All tests were 2-sided with significance level of 0.05.

RESULTS

- 37,054 patients met the study criteria. Most (82.5%) patients had an annual AMR ≥0.5 compared with only 17.5% of patients with a low AMR.
- The characteristics of patients with a low AMR compared with patients with a high AMR was as follows (**Table**):
 - Younger than patients with a high AMR (mean age, 30.1 years vs 35.2 years, respectively; *P*<0.001); and
 - More likely to be female (52.6% vs 57.2%, respectively; *P*<0.001).

Table. Patients Demographics

Characteristic	Low AMR, <0.5 n=6,479	High AMR, ≥0.5 n=30,575	All Patients N=37,054	*P* Value
Mean (SD) age, y	30.1 (18.3)	35.2 (20.5)	34.3 (20.2)	<0.001
5–11	1,466 (22.6)	6,811 (22.3)	8,277 (22.3)	
12–50	3,852 (59.5)	13,848 (45.3)	17,700 (47.8)	
51–64	1,161 (17.9)	9,916 (32.4)	11,077 (29.9)	
Female, n (%)	3,411 (52.6)	17,482 (57.2)	20,893 (56.4)	<0.001
Region, n (%)				0.004
Midwest	1,643 (25.4)	8,303 (27.2)	9,946 (26.8)	
Northeast	814 (12.6)	3,842 (12.6)	4,656 (12.6)	
South	2,985 (46.1)	13,949 (45.6)	16,934 (45.7)	
West	1,037 (16.0)	4,481 (14.7)	5,518 (14.9)	

AMR, asthma medication ratio; SD, standard deviation.

- Patients with a low AMR had higher rates of any poor asthma control event compared with those with a high AMR: 74.3% versus 26.9%, respectively (*P*<0.001; **Figure 2**).
 - ≥6 SABA fills: 49.2% versus 6.2%, respectively (*P*<0.001).
 - ≥2 OCS bursts: 32.0% versus 20.3%, respectively (*P*<0.001).
 - ED visits: 24.0% versus 5.1%, respectively (*P*<0.001).
 - Hospitalization rates: 6.8% versus 1.9%, respectively (*P*<0.001).

Figure 2. Poor Asthma Control Events Stratified by AMR

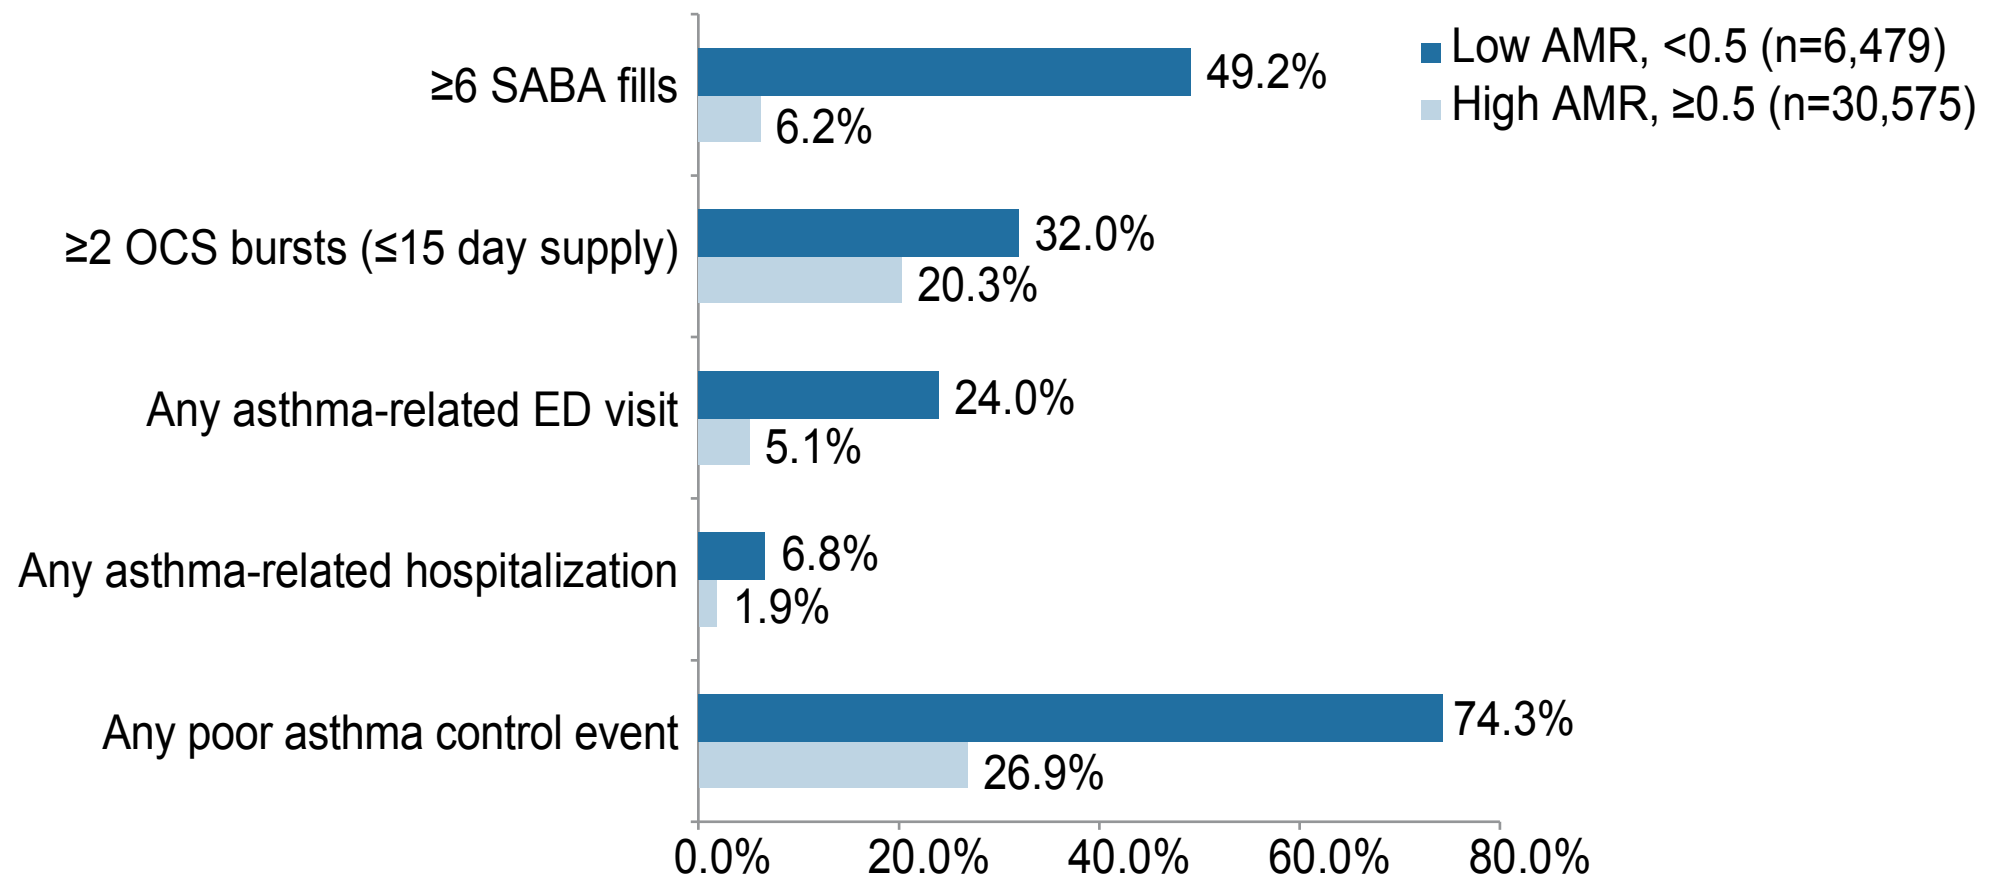
Low AMR, <0.5 (n=6,479)
High AMR, ≥0.5 (n=30,575)
≥6 SABA fills
49.2%
6.2%
≥2 OCS bursts (≤15 day supply)
32.0%
20.3%
Any asthma-related ED visit
24.0%
5.1%
Any asthma-related hospitalization
6.8%
1.9%
Any poor asthma control event
74.3%
26.9%
0.0%
20.0%
40.0%
60.0%
80.0%

AMR, asthma medication ratio; ED, emergency department; OCS, oral corticosteroid; SABA, short-acting beta agonist. *P*<0.001 for all comparisons.

- The number of asthma-related office visits with evaluation and management services was slightly higher for patients with a low AMR compared with those with a high AMR (1.9 [SD, 2.0] vs 1.8 [SD, 1.9], respectively; *P*<0.001).

CONCLUSIONS

- Patients with a low AMR (<0.5) had significantly worse asthma control by multiple measures including hospitalization and ED visits, OCS use, and SABA use.
- The number of asthma-related office visits also was slightly higher for patients with a low AMR.
- These findings support the use of AMR as a quality of care measurement for patients with persistent asthma.

REFERENCES

1. National Committee for Quality Assurance. HEDIS & performance measurement. http://www.ncqa.org/HEDISQualityMeasurement.aspx. Accessed March 10, 2014.
2. Schatz M, et al. *Chest*. 2006;130:43–50.

DISCLOSURES

Karina Raimundo, Paul G. Solari, and James L. Zazzali are employees of Genentech, Inc. Michael Broder and Eunice Chang are employees of the Partnership of Health and Analytic Research.

ACKNOWLEDGMENTS

This study was funded by Genentech, Inc., South San Francisco, CA. Medical editing support for this poster was provided by Linda Wagner of Excel Scientific Solutions and funded by Genentech, Inc.

Presented at the Academy of Managed Care Pharmacy 26th Annual Meeting & Expo, April 1–4, 2014, Tampa, FL

Copyright © 2014. All rights reserved. Contact Karina Raimundo for permission.